Neural dynamics of the speed-accuracy trade-off

P203

Dominic Standage[1], Da-Hui Wang[2] and Gunnar Blohm[1]

1 Department of Biomedical and Molecular Sciences / Centre for Neuroscience Studies, Queen's University, Canada
2 Department of Systems Science, Beijing Normal University, China

mailto: standage@queensu.ca

Introduction

Decisions are faster and less accurate when conditions favour speed (*vice versa* for accuracy)
The speed-accuracy trade-off (SAT) is a control mechanism for decision processing
See Standage et al, Front Neurosci, 2014

The SAT is well characterized by the principles of bounded integration
A higher decision bound favours accuracy at the expense of speed (and *vice versa*)
See Ratcliff and McKoon, Neural Computation, 2008

It is widely believed that the bound is implemented by a threshold rate of neural activity
... but the rate of decision-correlated neural activity is typically fixed at the time of choice commitment
e.g. Hanes and Schall, Science, 1996; Roitman and Shadlen, 2002; Ding and Gold, Cereb Cortex, 2012

Baseline rates are higher (lower) under speed (accuracy) conditions in several studies
e.g. Forstmann et al, PNAS, 2008; Ivanoff et al, PLoS ONE, 2008; Wenzlaff et al, J Neurosci, 2011; Heitz and Schall, 2012

It has been suggested that the threshold-baseline difference implements the bound-start difference
See Bogacz et al, TiNS, 2010

Neural models suggest that adjustments to baseline activity control the SAT by network dynamics
See Roxin and Ledberg, PLoS Comput Biol, 2008; Furman and Wang, Neuron, 2008; Standage et al, PLoS Comput Biol, 2013

In attractor models, spatially non-selective excitation changes the shape of the attractor landscape
These models suggest that the bound is not implemented by firing rates *per se*

Background

In abstract models, a common input to integrators controls the SAT by adjusting the bound-start difference

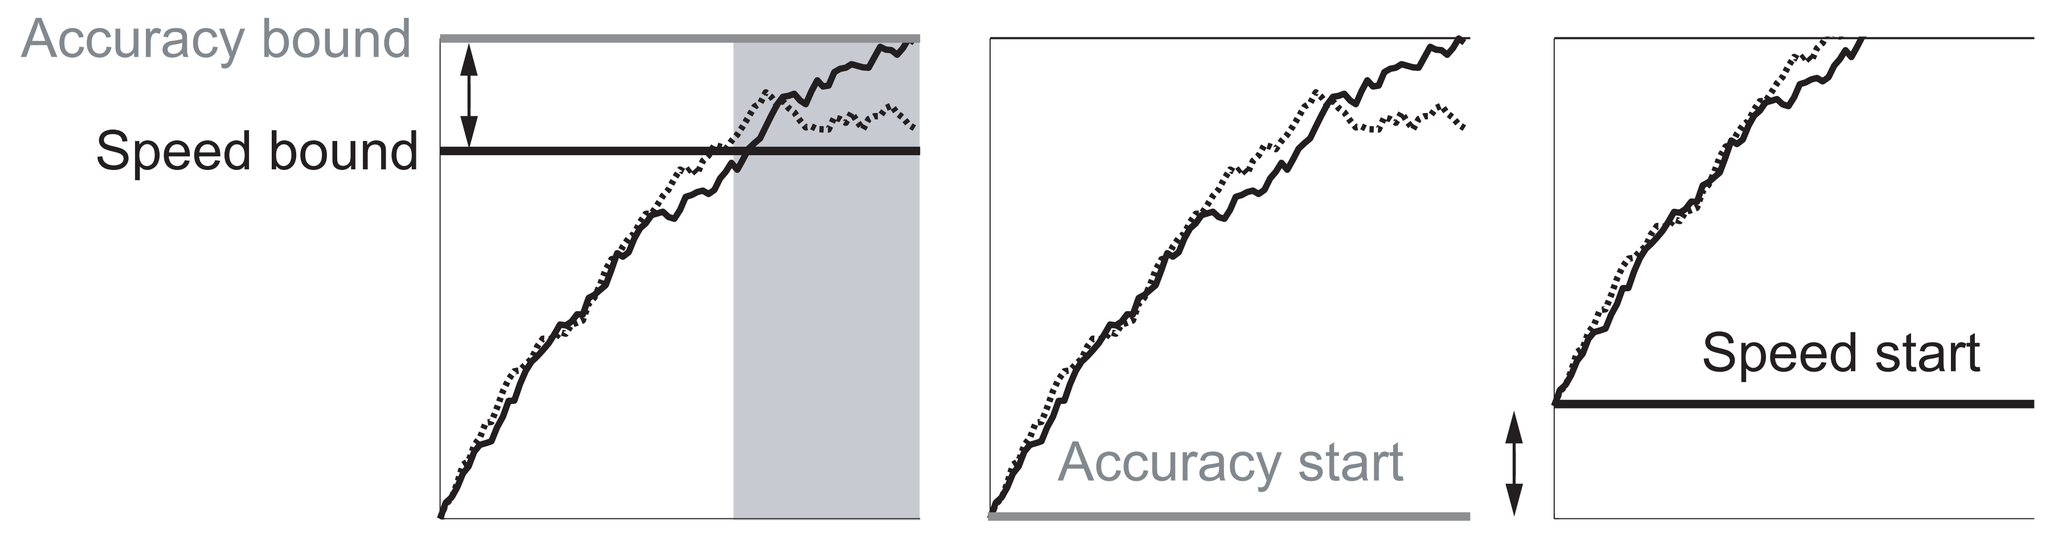
Accuracy bound
Speed bound
Speed start
Accuracy start
Time

See Bogacz et al, TiNS, 2010

Does the threshold-baseline difference implement the bound-start difference?

Methods

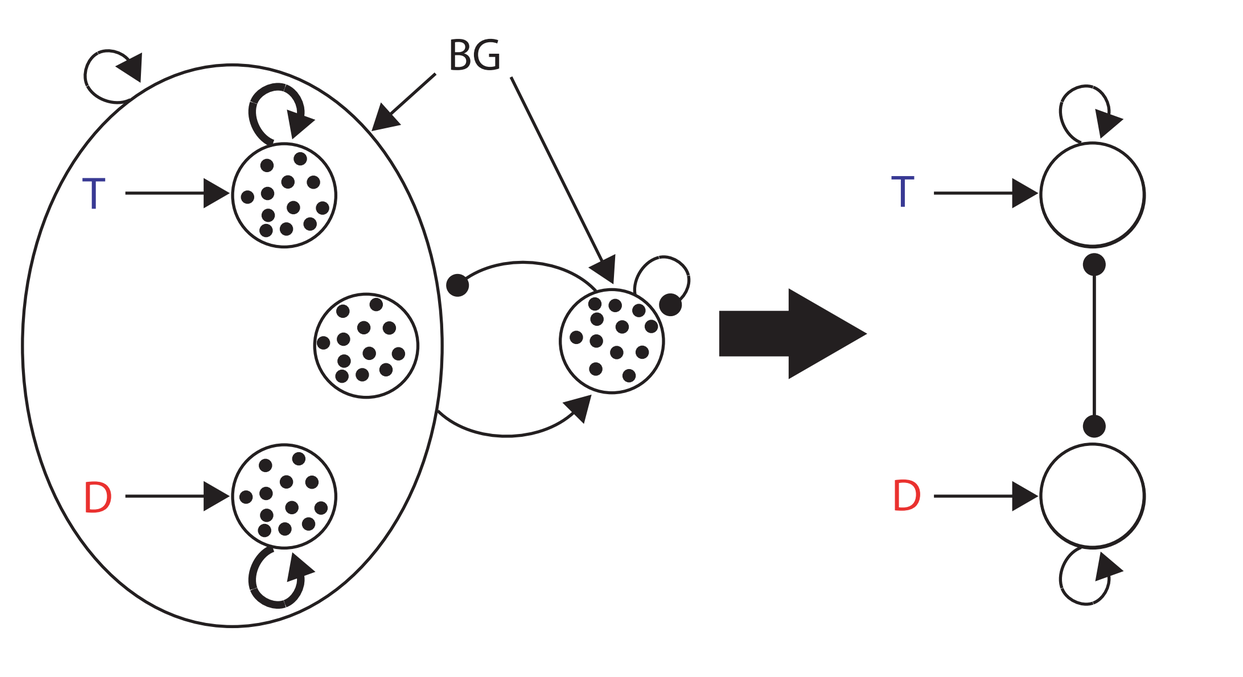
BG
T
D

Reduced biophysically-based network model
Wong and Wang J Neurosci, 2006

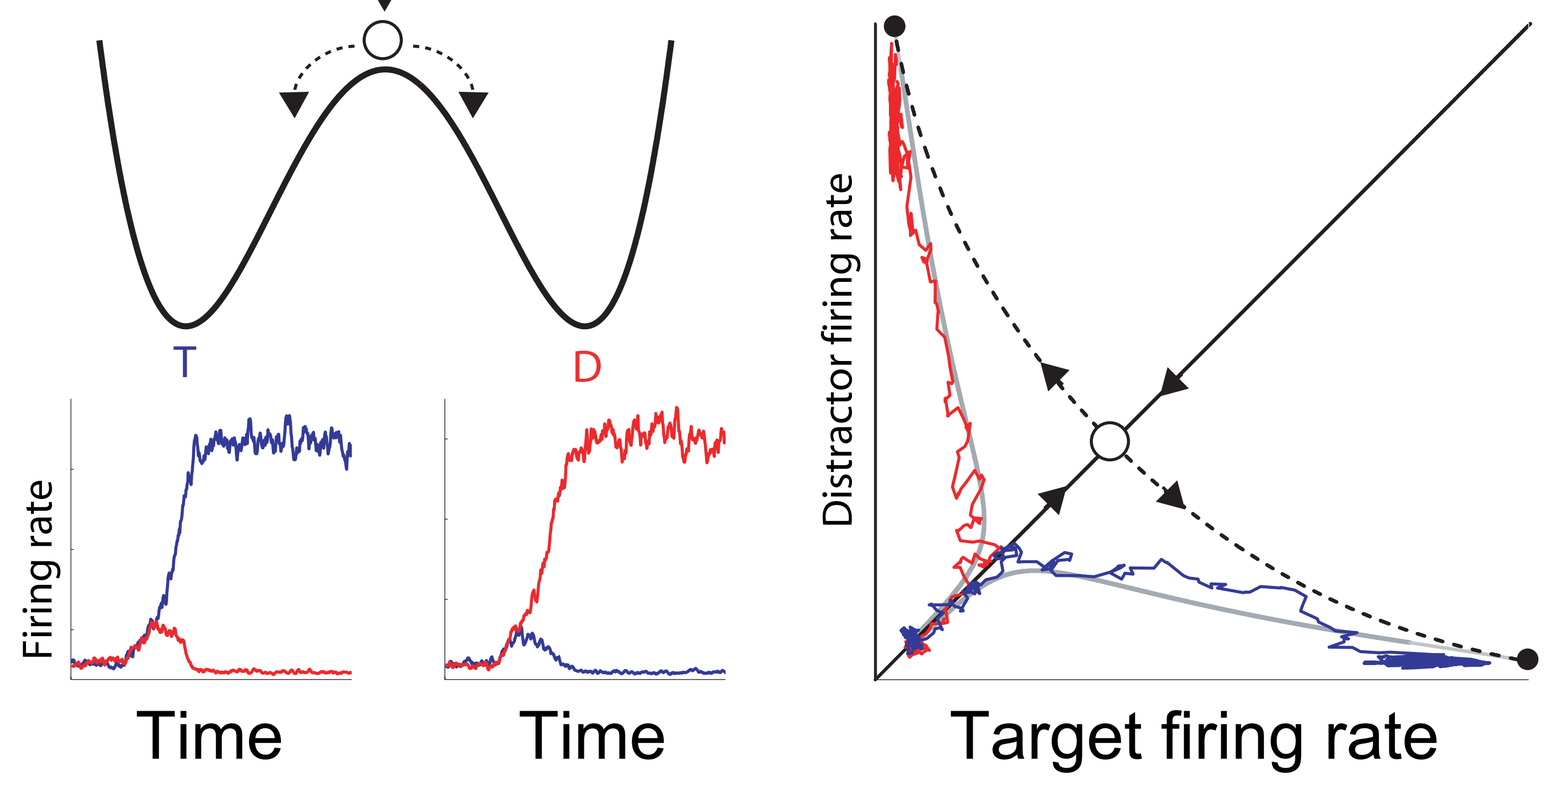
T
D
Firing rate
Time
Time
Distractor firing rate
Target firing rate

Network state is drawn toward the saddle point along the stable manifold (solid) and is repelled along the unstable manifold (dashed) toward an attractor state

Results

A common input to integrator populations controls the SAT

Simulated random dot motion task — Speed — Neutral — Accuracy

Stronger (weaker) non-selective input under speed (accuracy) conditions

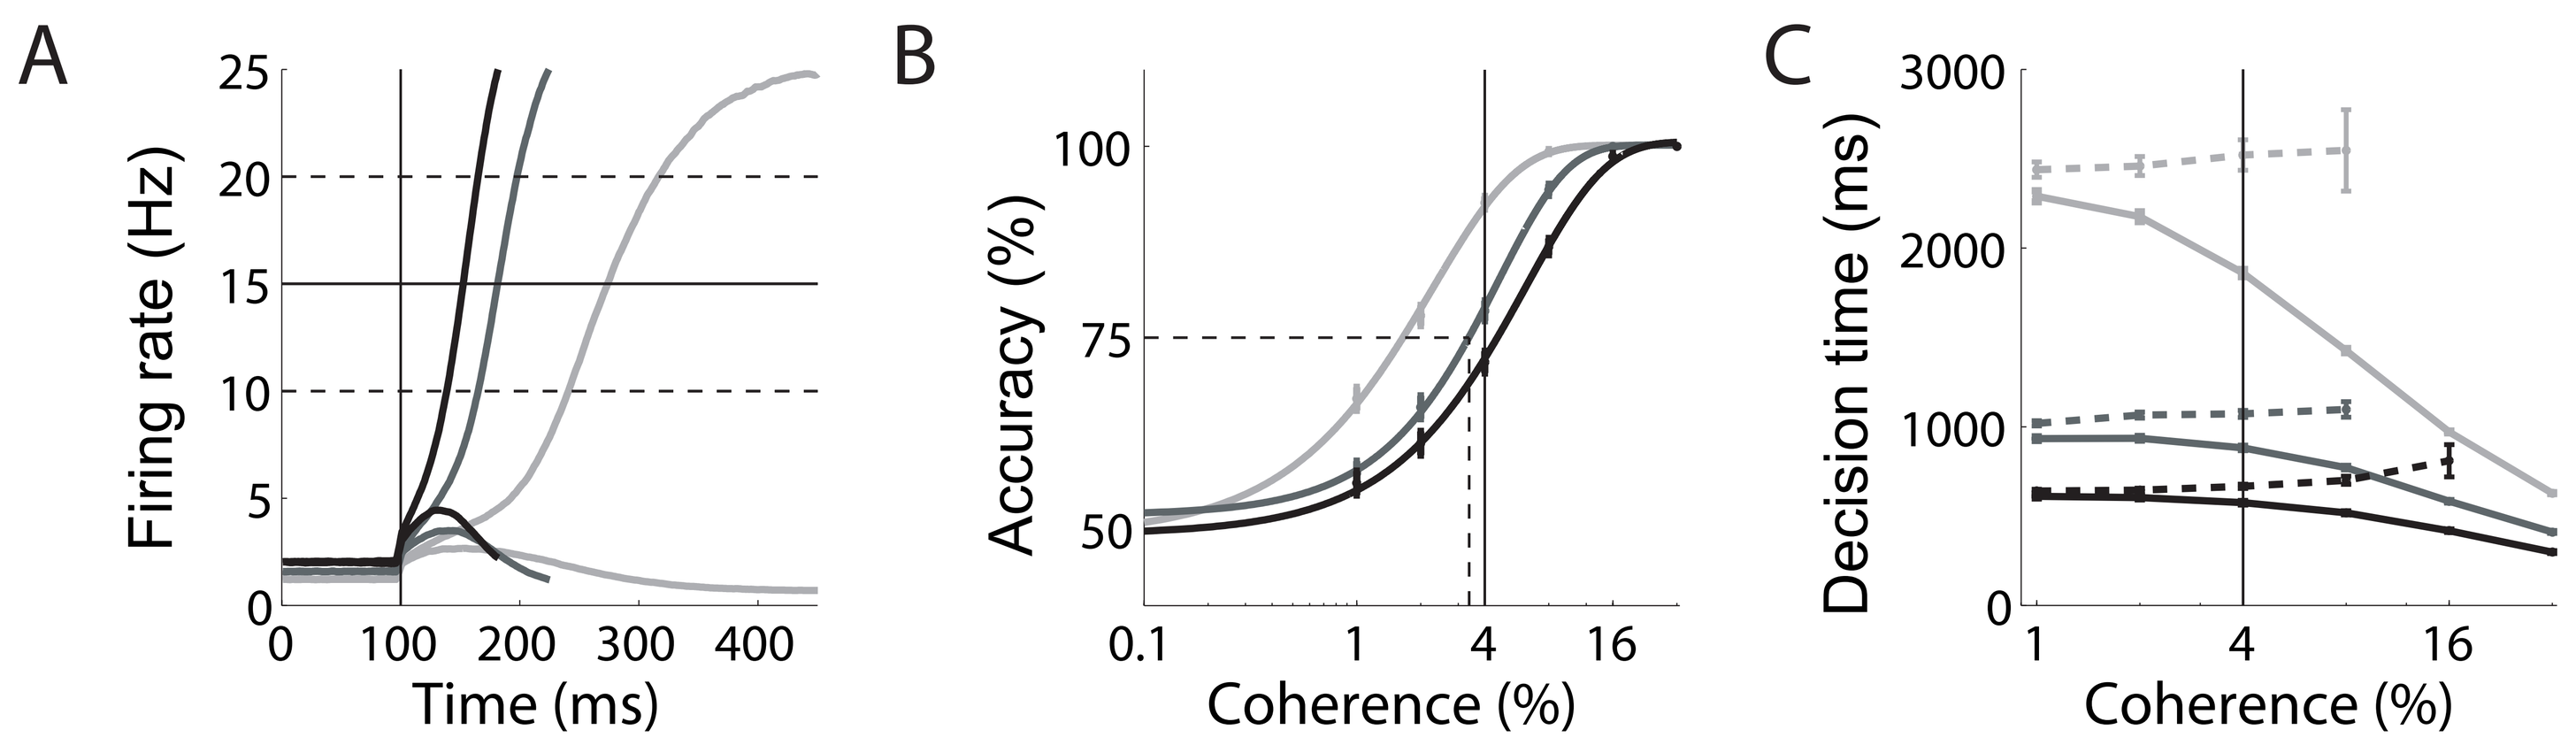
A
Firing rate (Hz)
Time (ms)
B
Accuracy (%)
Coherence (%)
C
Decision time (ms)
Coherence (%)

The threshold-baseline difference does not account for the SAT

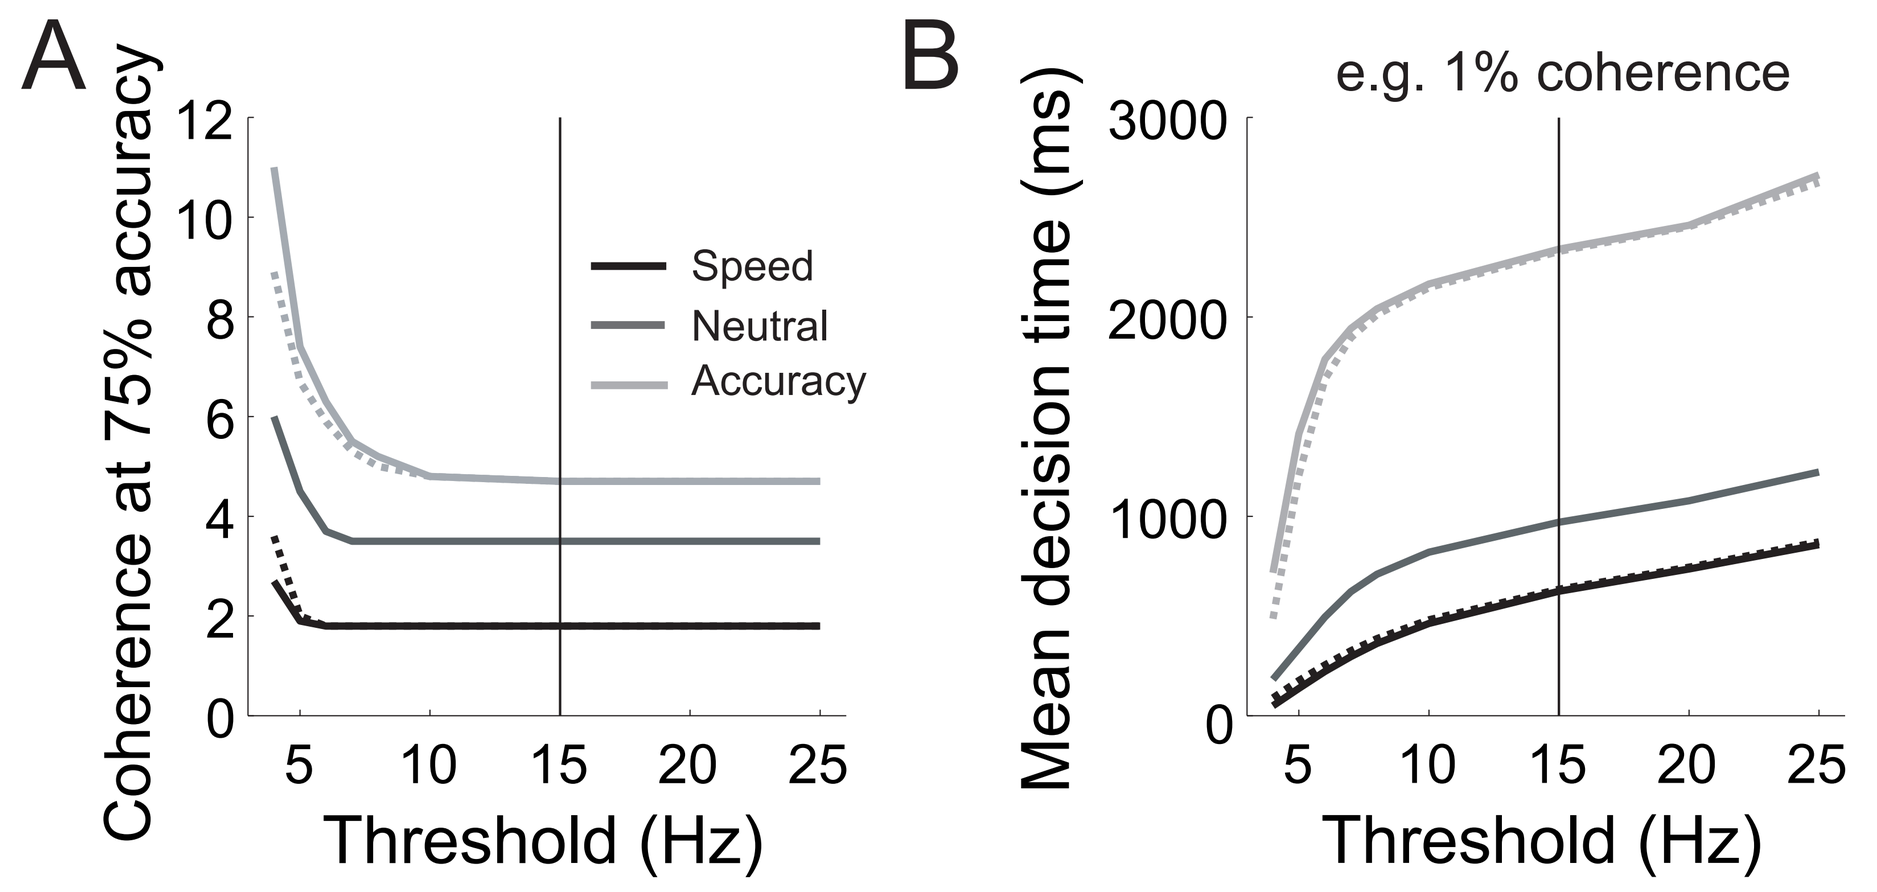
A
Coherence at 75% accuracy
Threshold (Hz)
Speed
Neutral
Accuracy
B
e.g. 1% coherence
Mean decision time (ms)
Threshold (Hz)

Adjusting the threshold to compensate for the change in baseline under each condition does not reverse the SAT

Changes in network dynamics control the SAT in the model

The time constant of the unstable manifold of the saddle decreases with increasing non-selective excitation
i.e. it decreases under speed conditions, like the bound of bounded integrator models

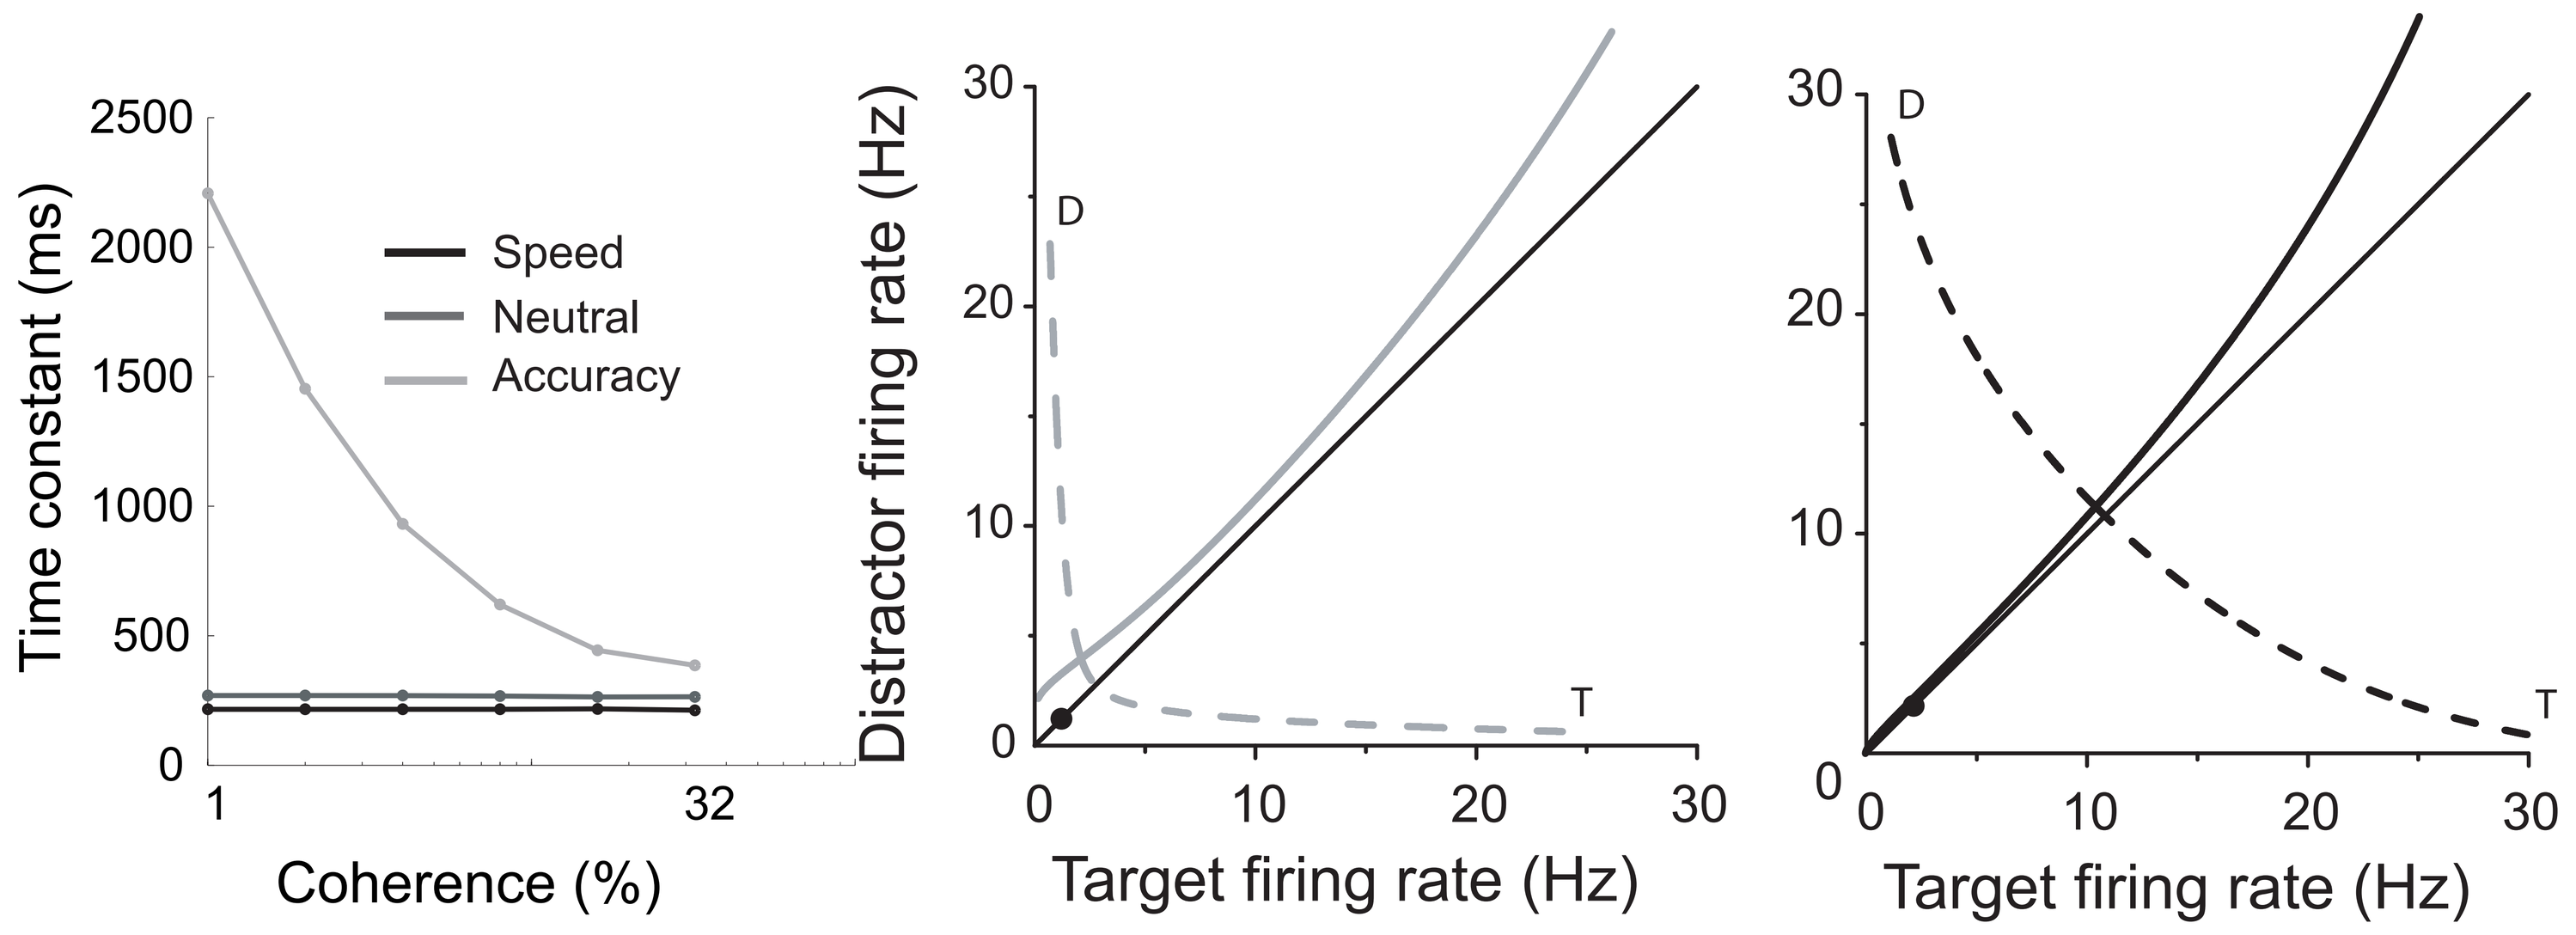
Time constant (ms)
Speed
Neutral
Accuracy
Coherence (%)
Distractor firing rate (Hz)
D
T
Target firing rate (Hz)
Target firing rate (Hz)

Stronger non-selective excitation pushes the stable manifold closer to the initial state
Errors occur when the state crosses the stable manifold, so accuracy is further reduced under speed conditions

Predictions for neural activity in SAT tasks

Firing rates will be higher under speed conditions at the time an ideal observer can discriminate target activity from distractor activity

... so will the difference between this rate and the baseline rate

Opposite to the principles of the threshold-baseline hypothesis

Standage, Wang and Blohm, in review

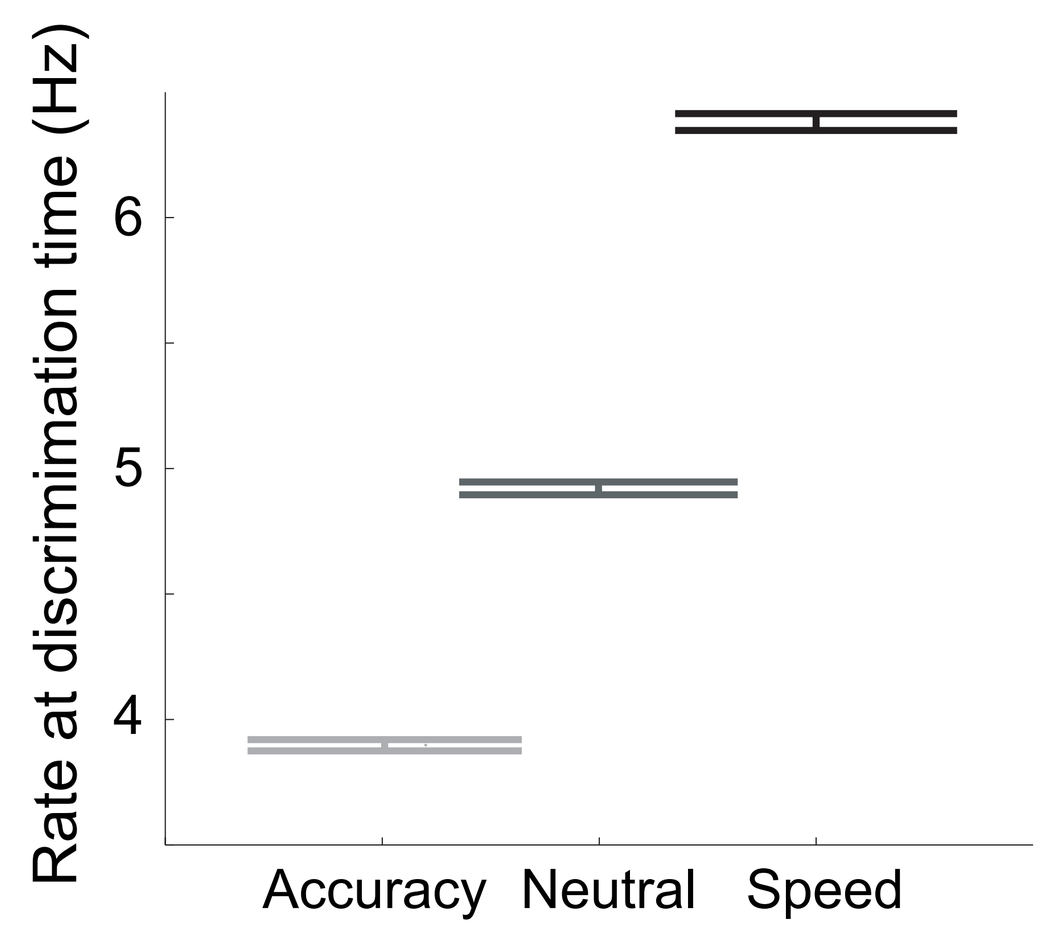
Rate at discrimination time (Hz)
Accuracy
Neutral
Speed

Heitz and Schall, Neuron, 2012

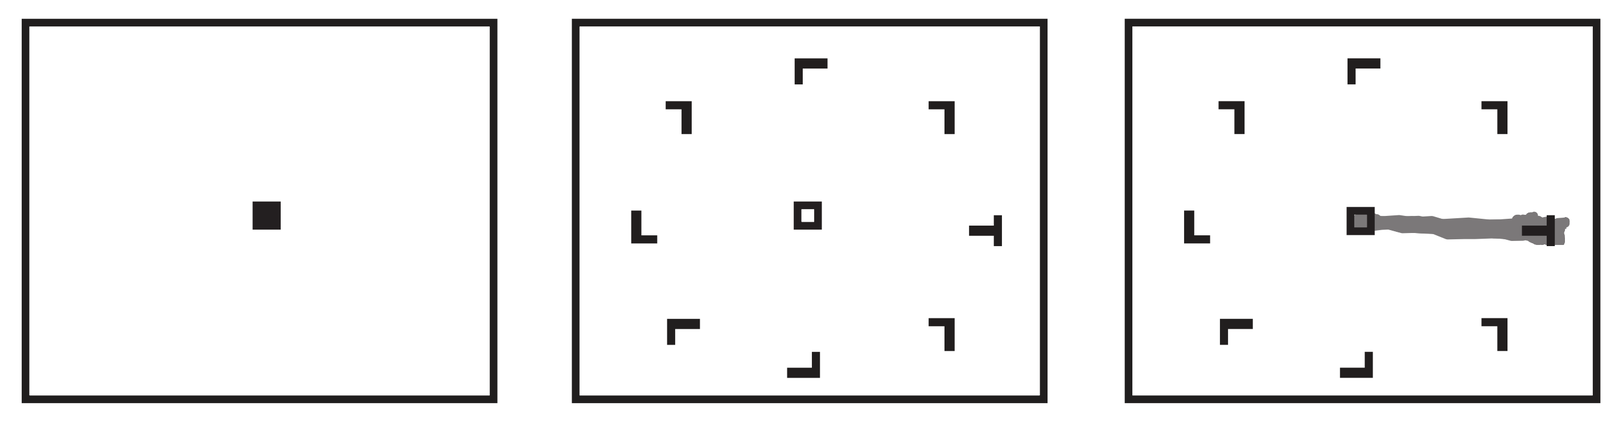

Monkeys performed an 8-choice visual search task under speed and accuracy conditions

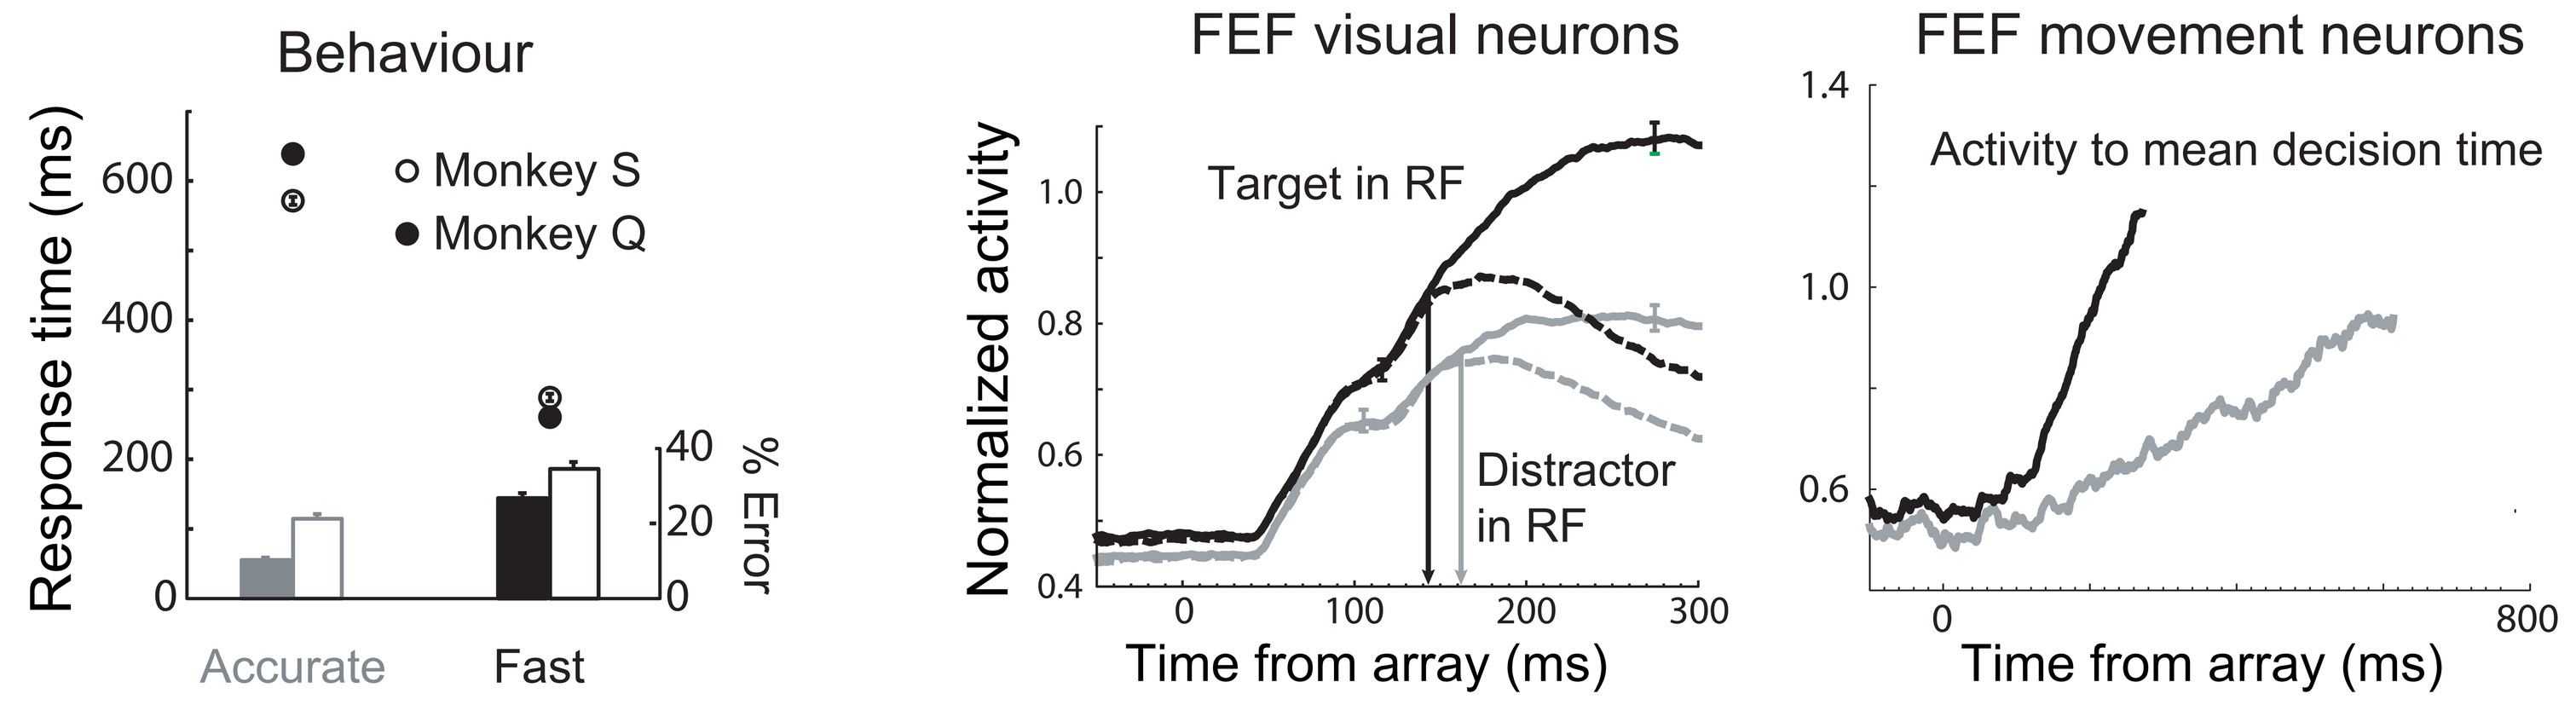
Behaviour
Response time (ms)
Monkey S
Monkey Q
% Error
Accurate
Fast
FEF visual neurons
Normalized activity
Target in RF
Distractor in RF
Time from array (ms)
FEF movement neurons
Activity to mean decision time
Time from array (ms)

Under speed conditions (vice versa for accuracy)
Visual neurons showed (1) higher baseline, (2) higher response magnitude and (3) earlier target-in / target-out separation
Movement neurons showed (4) higher baseline, (5) higher rate of rise and (6) higher peak rate

Coupled-circuit FEF model with synaptic resolution

FEF movement neurons are hypothesized to integrate the activity of evidence-encoding FEF visual neurons
See Purcell et al, J Neurosci, 2012

Background actrivity by noisy current injection
See Destexhe et al, Neurosci, 2001

Visual stimuli and a non-selective control signal by Poisson spike trains

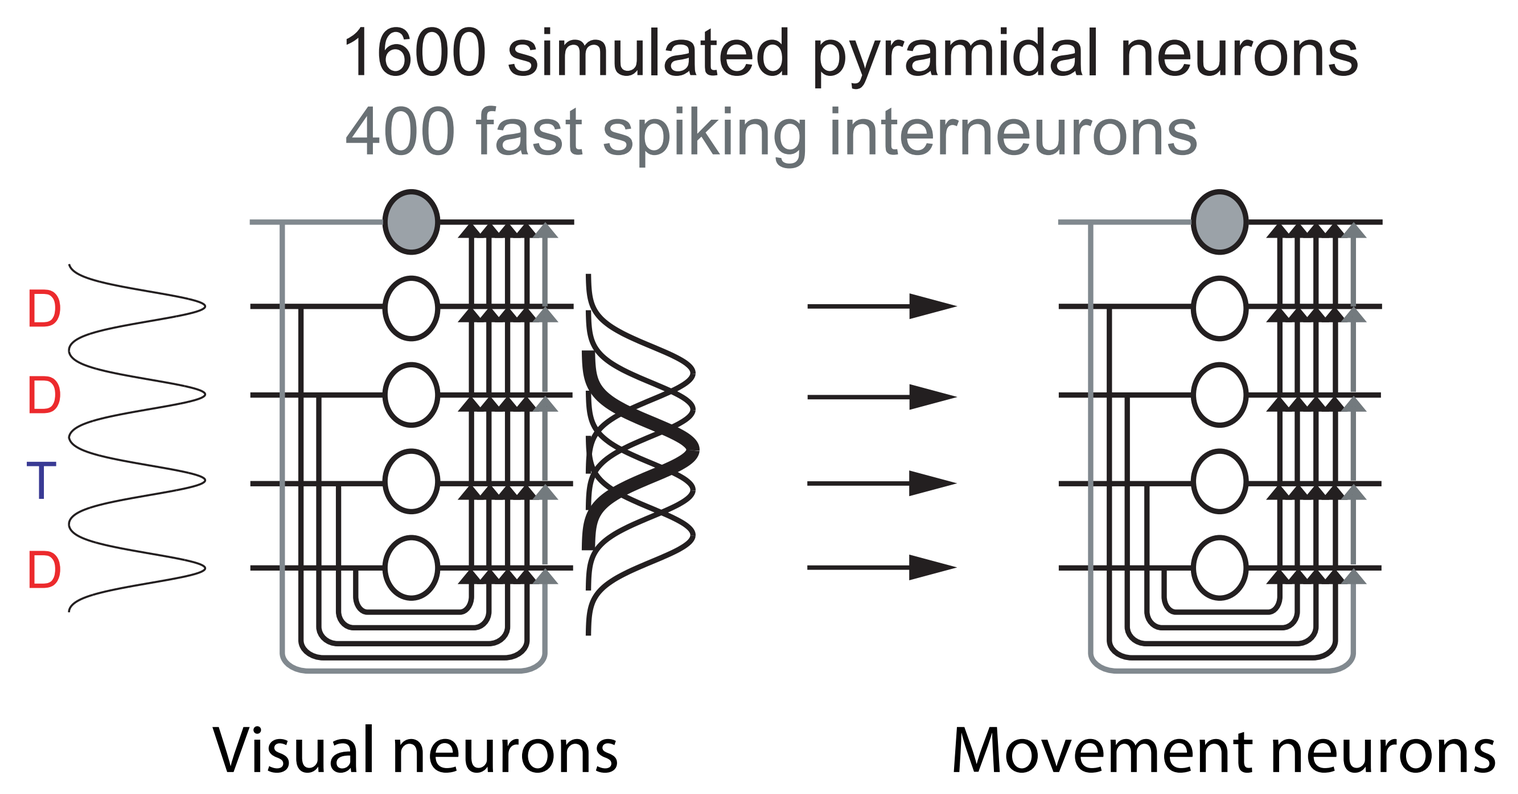
1600 simulated pyramidal neurons
400 fast spiking interneurons
D
D
T
D
Visual neurons
Movement neurons

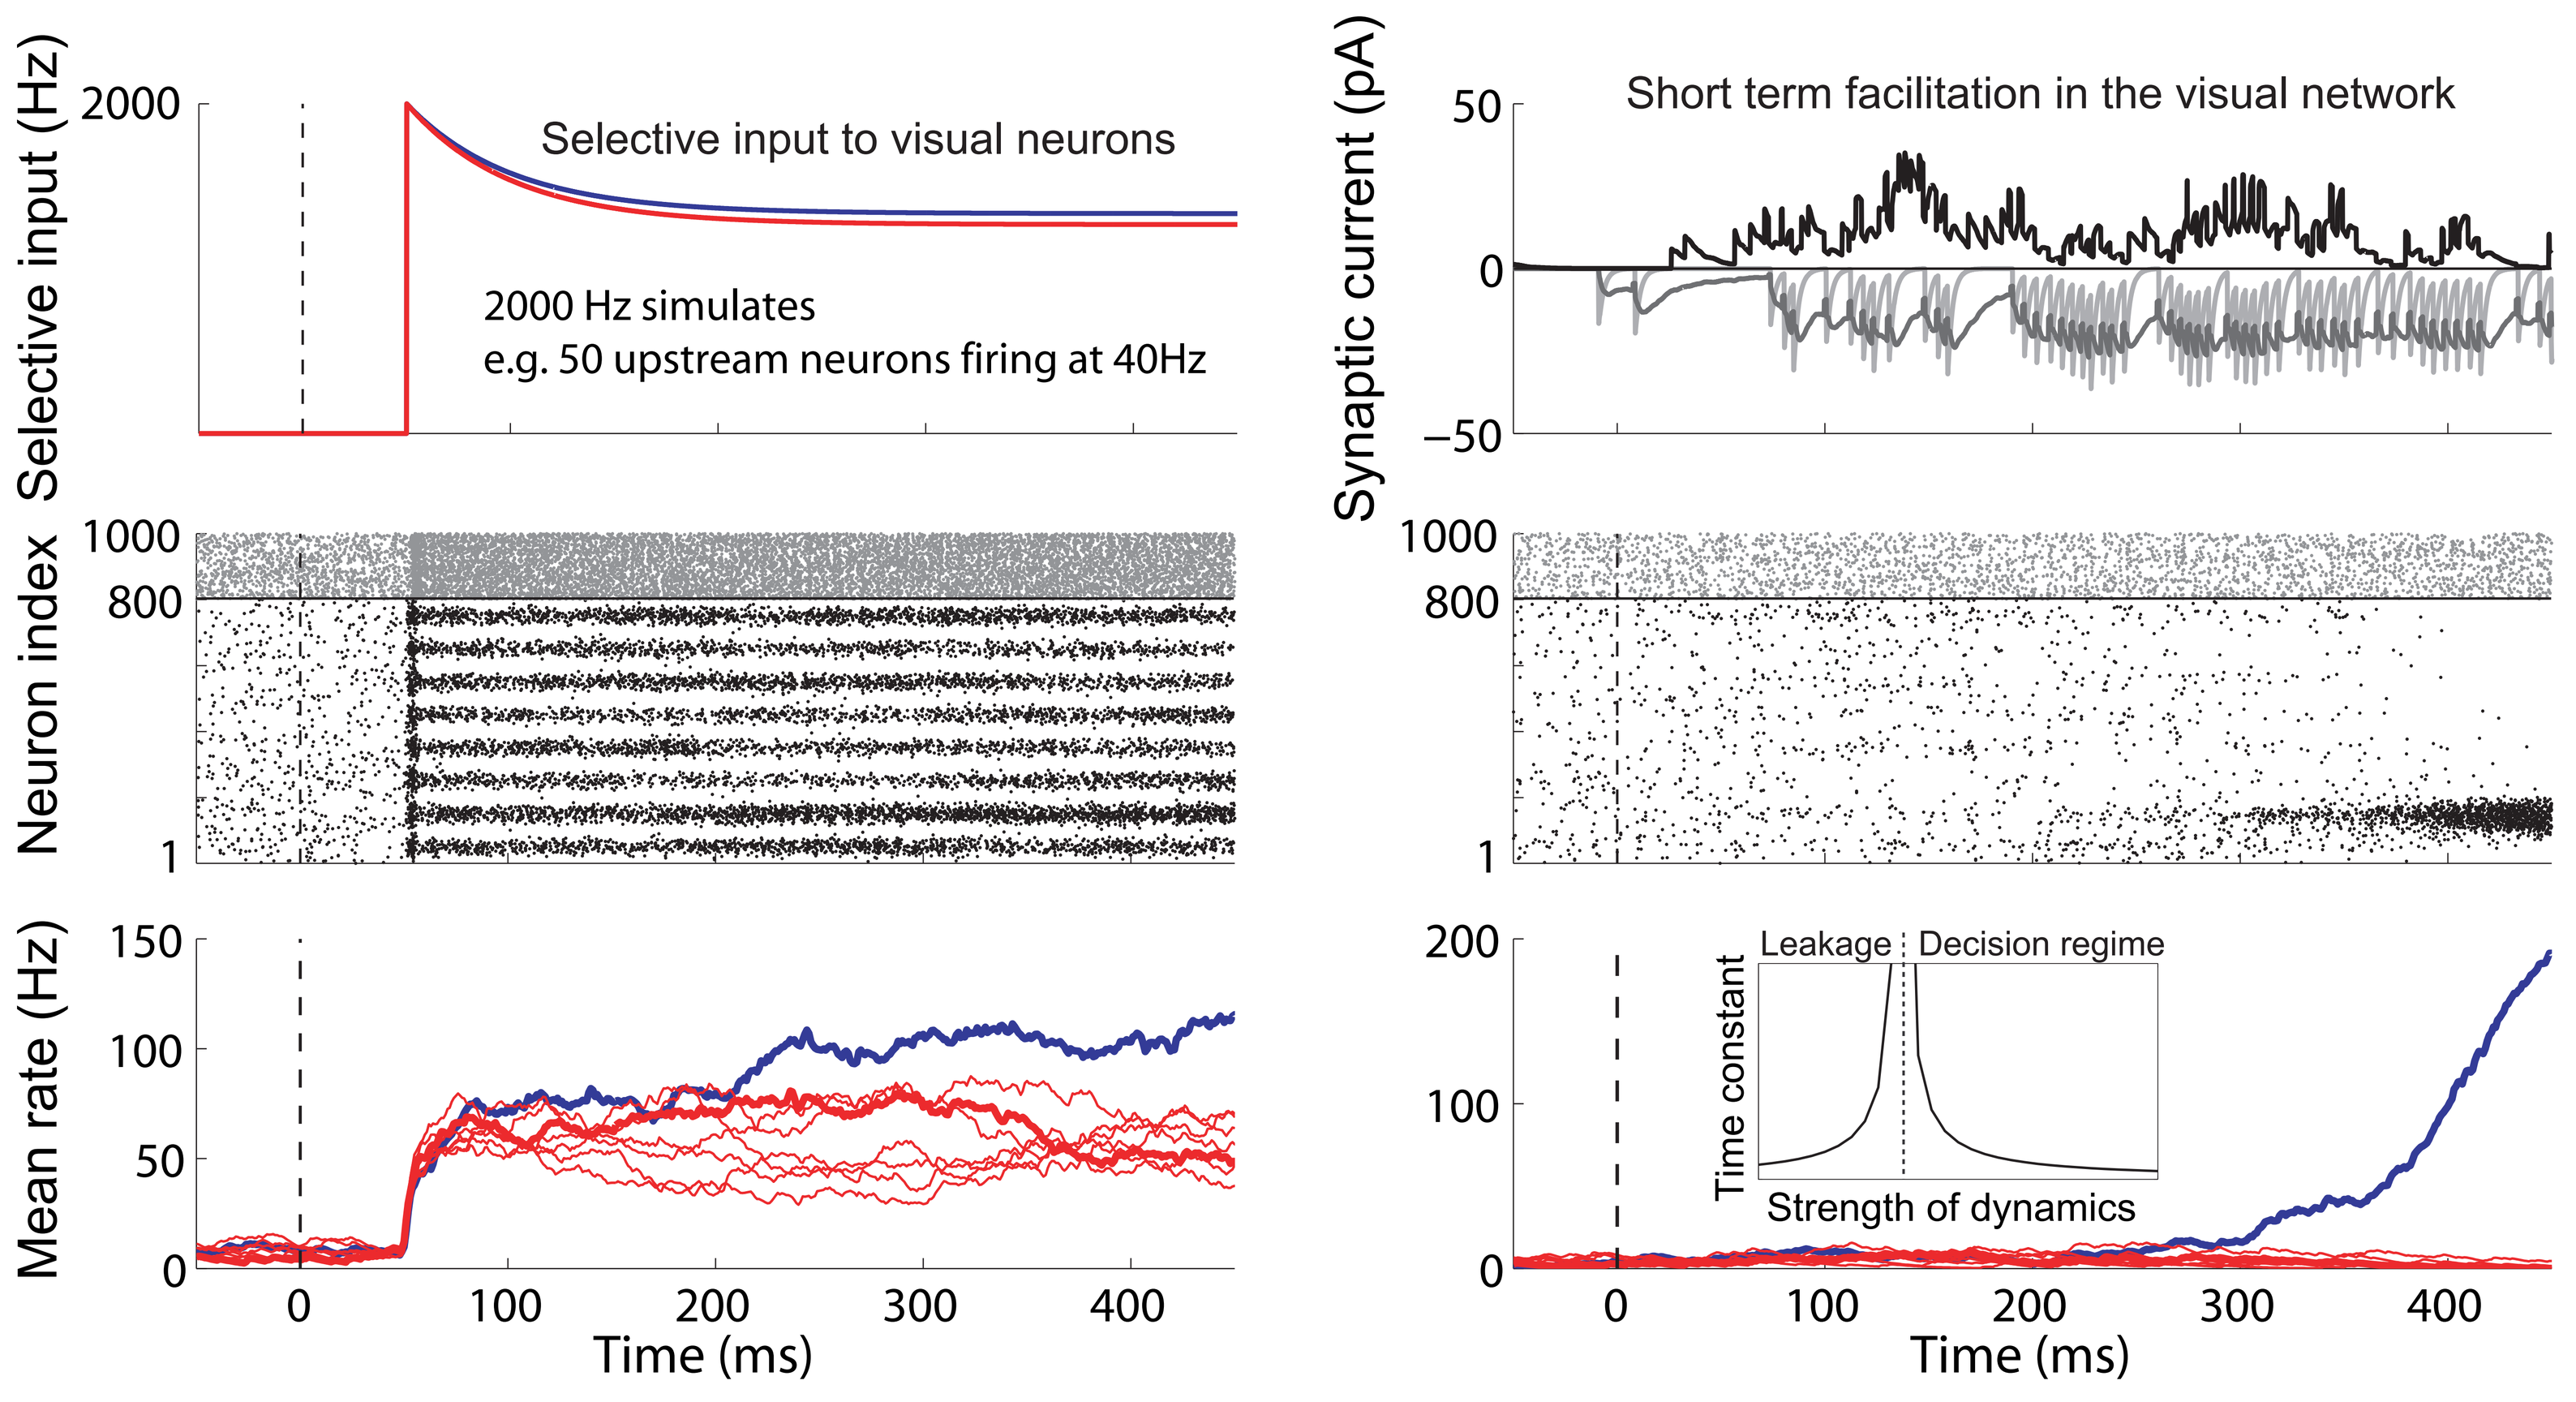
Selective input (Hz)
Selective input to visual neurons
2000 Hz simulates
e.g. 50 upstream neurons firing at 40Hz
Neuron index
Mean rate (Hz)
Time (ms)
Synaptic current (pA)
Short term facilitation in the visual network
Leakage
Decision regime
Time constant
Strength of dynamics
Time (ms)

Visual neurons → Movement neurons

Preliminary results are encouraging

Qualitatively consistent with the data by Heitz and Schall (2012)
... plenty of room for quantitative improvement

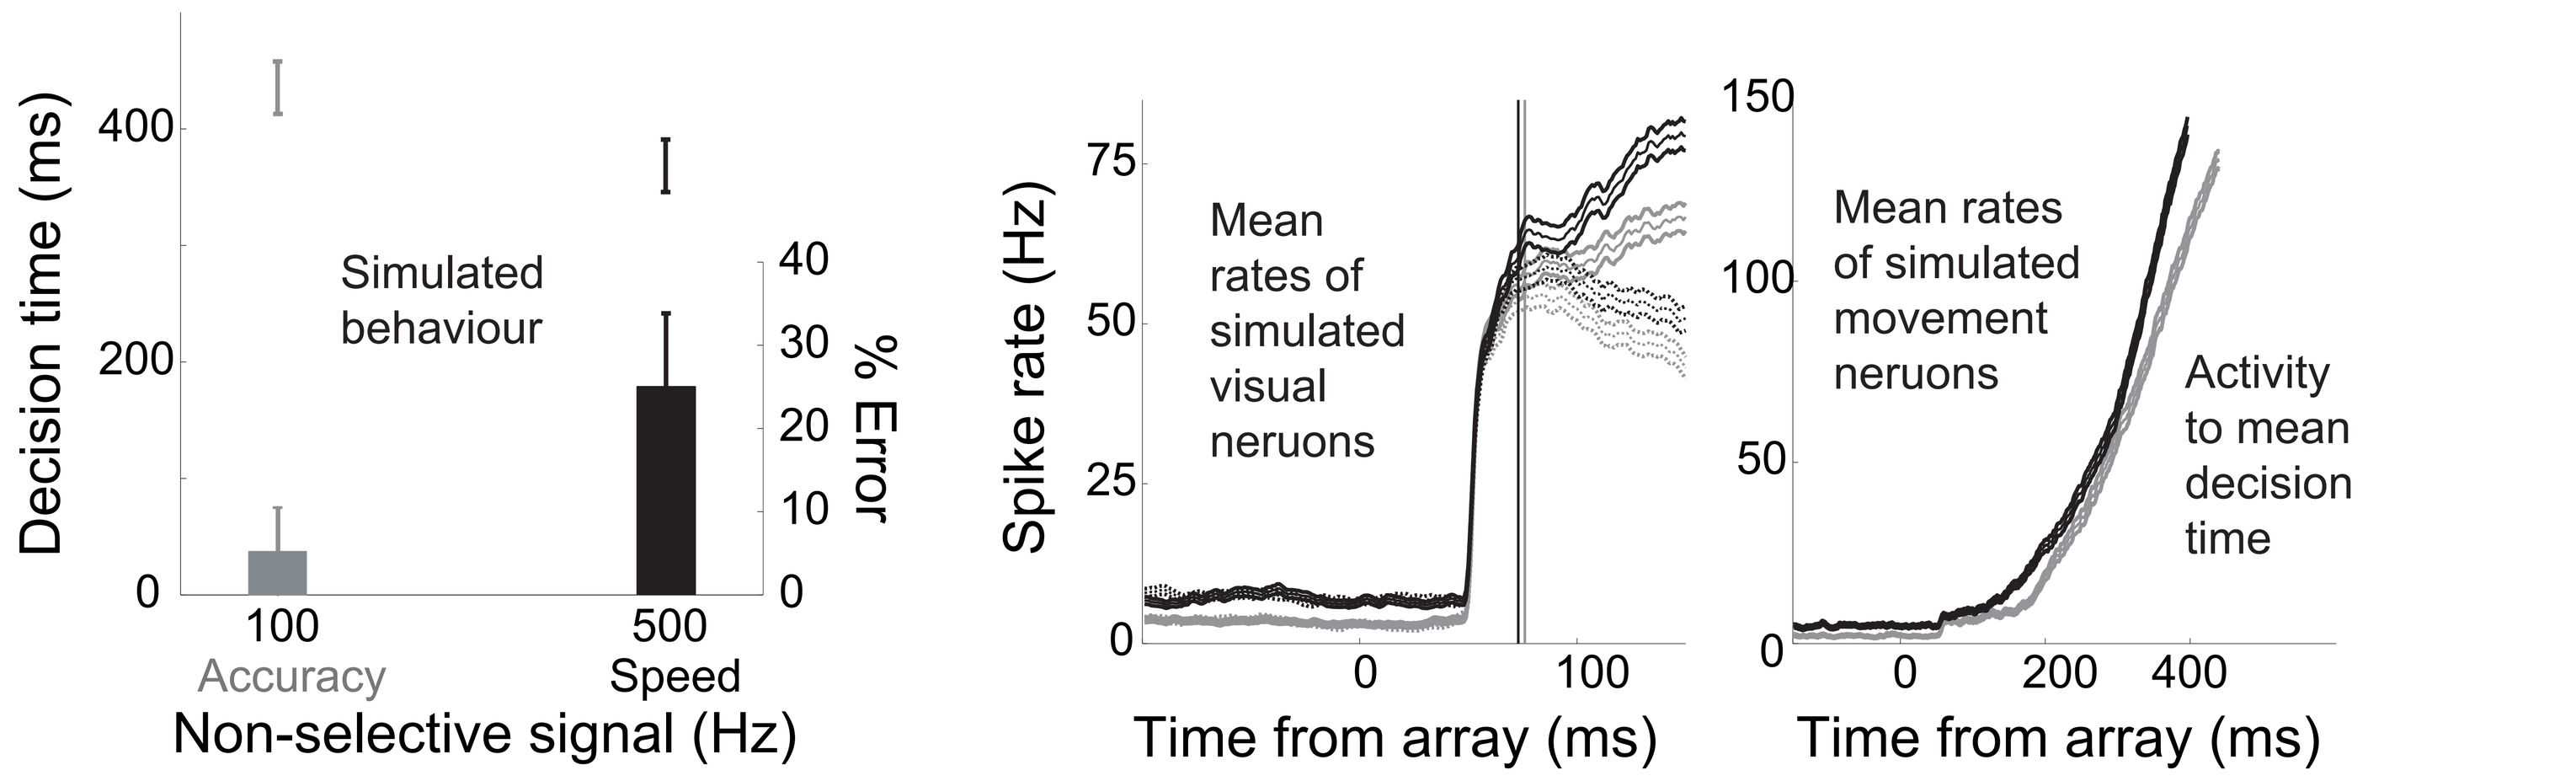
Decision time (ms)
Simulated behaviour
% Error
Accuracy
Speed
Non-selective signal (Hz)
Spike rate (Hz)
Mean rates of simulated visual neruons
Time from array (ms)
Mean rates of simulated movement neruons
Activity to mean decision time
Time from array (ms)

Conclusions and future work

Our study supports the hypothesis that a cognitive signal controls the SAT by projecting uniformly to integrator populations
See Standage et al, Front Neurosci, 2014

The resulting changes to the threshold-baseline difference may be epiphenomenal
Network dynamics control the SAT in attractor models

The rate of decision-correlated activity at the time of choice commitment is qualitatively different than the bound of bounded integrator models
The latter may be implemented by the effective time constant of decision circuity, not firing rates *per se*

Can all of the modulatory effects shown by Heitz and Schall (2012) be explained by changes to network dynamics engenered by a spatially non-selective cognitive signal?

Supported by the Canadian Institutes of Health Research